Promoter Control of Gene Expression in Adenovectors Delivered to the Vestibular System

Hinrich Staecker[1], Mark Praetorius[2], Chi Hsu[3], Kim Baker[4] and Douglas E. Brough[3]
[1]Department of Otolaryngology, University of Kansas School of Medicine, Kansas City KA
[2]Department of Otolaryngology, University of Heidelberg, Heidelberg Germany
[3]Research Department, Vector Sciences, GenVec Inc., Gaithersburg MD
[4]Department of Otolaryngology Head and Neck Surgery, University of Maryland, Baltimore MD

Abstract

Delivery of the atonal homologue math1 has been demonstrated to result in the generation of hair cells after aminoglycoside damage. To study the dynamics of hair cell regeneration, we treated adult mouse macular organ cultures with neomycin followed by an advanced generation adenovector carrying math1 driven by the human CMV promoter, the CMV enhancer chicken beta actin promoter or the glial fibrillary acidic protein (GFAP) promoter. These promoters differ both in strength of expression, expression kinetics and cell specificity. Total dose of vector was varied as was the time post neomycin treatment that vector was administered to the explants. At 5 day intervals explants were fixed and either whole mount stained for myosin VII or serially sectioned and stained for myosin VII. Total hair cell counts were obtained for each culture condition. The choice of promoter was found to significantly affect the math1 expression levels and the effect of math1 on damaged tissue. *In vitro* restoration of hair cells could be seen by 5 days post math1 delivery and *in vivo* restoration of hair cells with the GFAP promoter construct was effective.

Introduction

Delivery of atoh1 to the inner ear triggering the transdifferentiation of supporting cells to hair cells has been suggested as a possible treatment for loss of sensory hair cells. In the design of such a treatment for hearing loss and balance disorders it may be beneficial to specify the level of expression, duration of expression and localization of transgene expression. To begin the design of adenovectors for delivering genes to the vestibular system we analyzed the impact of different promoter combinations and delivery time course. The basic construction of Ad5 serotype vectors involves deleting different portions of the adenovirus genome necessary for virus replication placing these genes into the genome of cell lines used to propagate the vectors. It has been previously reported that mutations and partial deletions of the adenovector polymerase gene or deletion of the adenovirus E4 region, in addition to deletion of the E1 and E3 regions, result in vectors that have different levels and durations of transgene expression in specific tissues. In addition, the promoter used to drive the delivered transgene can influence the expression of the transgene in a tissue specific fashion. Also, the selection of promoters impacted the location, duration and level of gene expression. Delivery of atoh1 using the tissue selective GFAP promoter induced the most efficient recovery of hair cells. These results suggest that vector designs including a tissue selective promoter driving atoh1 with and E1/E3/E4 deleted vector is likely to have both safety and efficacy advantages for the treatment of balance disorders.

Atonal Mediated Hair Cell Regeneration

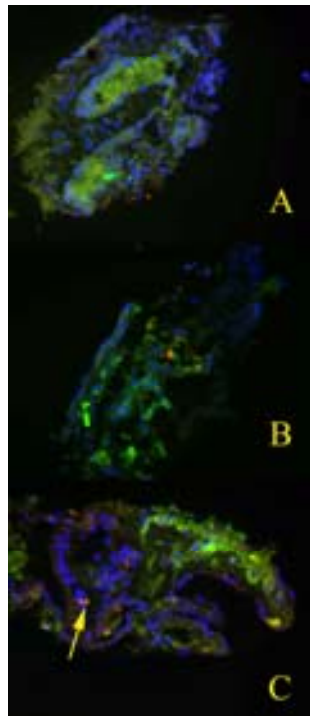
A
B
C

Figure 1: Time course of atoh1 mediated hair cell replacement *in vitro*: Macular organ explants were treated with neomycin 10-3 M for 48 hours and then transfected with Ad.atoh1.gfp.11d, a vector that expressed both gfp and atoh1. At 2 days post vector delivery gfp expression (Green) can be seen in the explant (A). Nuclei are labeled with DAPI (Blue). Five days after vector delivery the first signs of myosin VII expression (Red) are noted (B) with increasing numbers of myosin VII positive cells being seen after 10 days (C).

Transgene Expression from Different Promoters

Figure 2: Activity of different constitutively active promoters in inner ear tissue was assessed by using luciferase expressing vectors in macular organ cultures and measuring the average luciferase activity per µg of extracted protein over time. Significant differences in expression were noted when comparing promoter types. Use of the human CMV promoter resulted in a steady level of luciferase production (AdL.11D) that declined slowly after 1 week. The ubiquitin promoter demonstrated a slow decline of luciferase production over the 2 week study period (AdUb.L.11D). Expression of luciferase driven by the hybrid CMV enhancer / chicken beta actin promoter resulted in a steady increase in luciferase production (AdcBA.L.11.D).

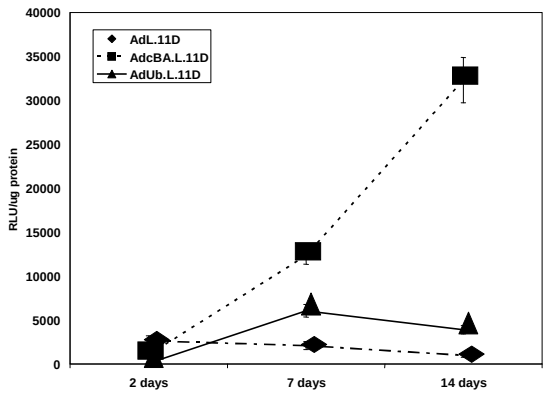
AdL.11D
AdcBA.L.11D
AdUb.L.11D
RLU/ug protein
40000
35000
30000
25000
20000
15000
10000
5000
0
2 days
7 days
14 days

GFAP Localization in the Inner Ear

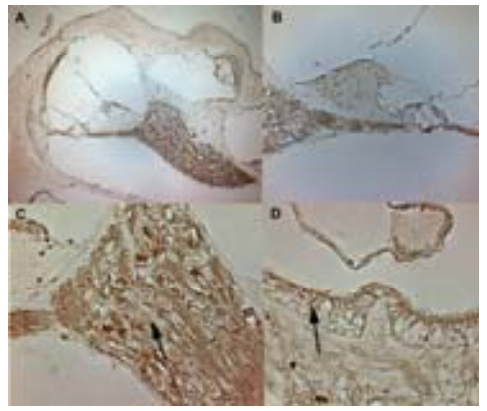
A
B
C
D

Figure 3: Immunohistochemistry for GFAP demonstrates that distribution of immunoreactivity closely parallels the distribution of GFP driven by the GFAP promoter

A. Low Power Cochlea, B. Organ of Corti (arrow on supporting cell), C. Spiral Ganglion with ganglion cells not labelling (arrow), D. Vestibular End Organ with arrow on Supporting Cell

Figure 4: Expression of GFP driven by a tissue specific promoter (GFAP promoter) results in labeling of supporting cells in the organ of Corti, vestibular supporting cells and glial cells in the spiral ganglion.

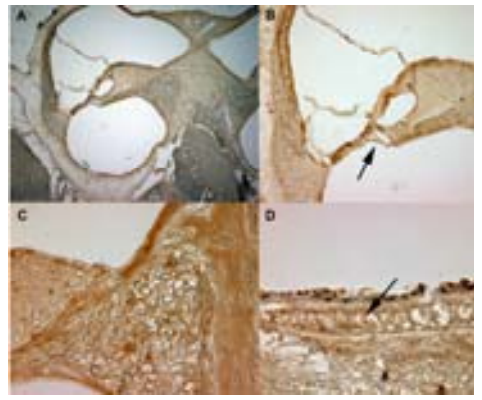
A
B
C
D

Regeneration of Hair Cells with a GFAP Promoter Driven Atonal Adenovector

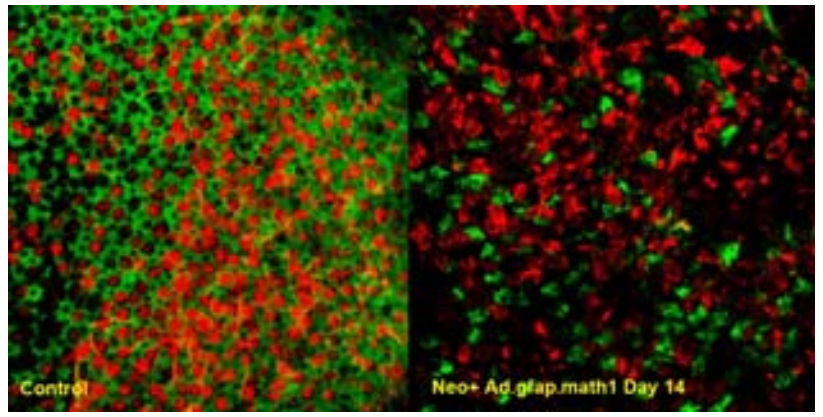
Control
Neo+ Ad.gfap.math1 Day 14

Figure 5: Whole mount macular organ cultures maintained *in vitro* for 16 days and immunostained for the presence of myosin VII (Red) demonstrated a dense population of hair cells (A). Explants treated with neomycin did not demonstrate significant recovery of hair cells (see Fig 6). Explants treated with neomycin followed by atoh1 driven by a GFAP promoter after neomycin treatment resulted in regeneration of hair cells (B).

Promoter Choice for Atonal Expression Makes a Difference

Figure 6: Effect of promoters on hair cell regeneration *in vitro*: Hair cell counts for macular organ cultures treated with neomycin followed by atoh1 delivered by adenovectors with hCMV, CBA and GFAP promoters. Controls consisted of untreated macular organ cultures, macular organ cultures treated with neomycin for 2 days and then fixed or 2 days for neomycin treatment followed by 14 days of culture. A third control consisted of macular organ cultures treated with neomycin followed by exposure to an adenovector expressing gfp (Adf.11D). Hair cell counts were determined by counting total number of myosin VII positive cells in the explant (n=5 each condition). Neomycin treated explants or neomycin + Ad.11d treated explants showed only minimal presence of myosin VII positive cells. Recovery of myosin VII positive cells was optimal using the GFAP promoter construct. The cBA promoter, which yielded the highest expression level of transgene (Fig 2) showed the lowest level of recovery.

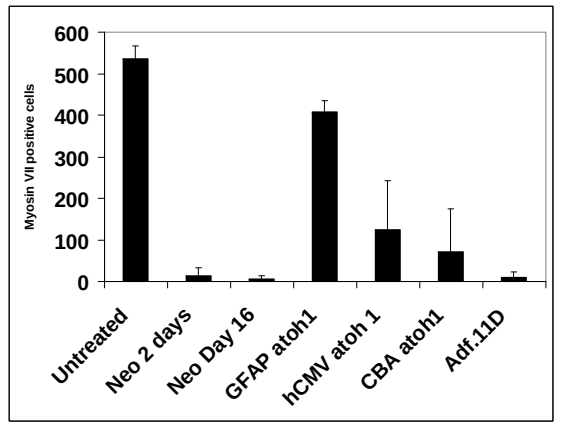
Myosin VII positive cells
600
500
400
300
200
100
0
Untreated
Neo 2 days
Neo Day 16
GFAP atoh1
hCMV atoh 1
CBA atoh1
Adf.11D

GFAP Expressed Atonal Mediated Hair Cell Regeneration *In Vivo*

Figure 7: Delivery of atoh1 after delay *in vivo*: Adult mice were treated with aminoglycoside as previously described and allowed to recover for 10 days. 1 x 10 ^{8} pu of Ad.GFAP.atoh1 was then injected into the posterior semicircular canal. Animals were allowed to recover for 1 month and then processed for myosin VII immunohistochemistry. Controls (A) show a healthy population of hair cells in the macular organs. Aminoglycoside treated animals show minimal survival of hair cells (B). Aminoglycoside animals treated with atoh1 demonstrated recovery of a population of hair cells (C) that are innervated (D).

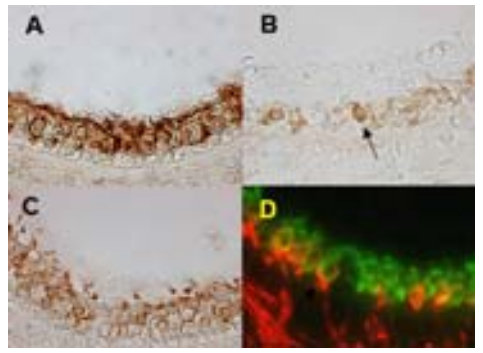
A
B
C
D

Conclusions:

1) Modification of promoters results in different expression levels and differential regenerative response to atoh1

2) Delivery of atoh1 using a supporting cell specific promoter is effective

Supported by NIDCD DC 07772