ADENOVECTOR-BASED MALARIA VACCINE INDUCES STRONG T-CELL AND ANTIBODY RESPONSES AGAINST MULTIPLE PRE-ERYTHROCYTIC-STAGE ANTIGENS

Joseph T. Bruder[1], Elena Semenova[1], Charlie Thomas[1], Duncan McVey[1], Svetlana Konovalova[1], Keith Limbach[2], Maureen E. Stefaniak[2], Noelle B. Patterson[2], Joseph J. Campo[2], Sheng Li[3], C. Richter King[1], Denise L. Doolan[2]

[1]GenVec, Inc., 65 West Watkins Mill Road, Gaithersburg, MD20878; [2]Naval Medical Research Center, Malaria Program, 503 Robert Grant Avenue, Silver Spring, MD 20910-7500; [3] The PATH Malaria Vaccine Initiative, 7500 Old Georgetown Road, 12th Floor, Bethesda, MD 20814

MALARIA – A World Health Problem

>1 million deaths/year
No licensed malaria vaccine
Drug resistance spreading

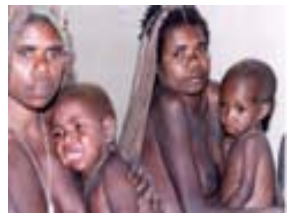

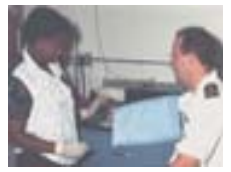

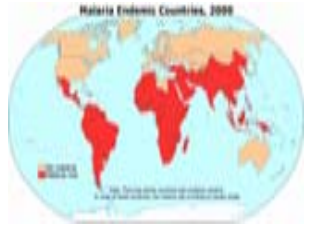
Malaria Endemic Countries, 2000

Product Opportunities

- Military Vaccine
- Travelers Vaccine
- Vaccine for Developing World

Malaria is the most important parasitic disease of man, exacting an estimated toll of greater than one million deaths and 350-500 million clinical cases per year. The feasibility of a malaria vaccine is supported by experimental data demonstrating protective immunity induced by exposure to the intact parasite. Immunization of animals or human volunteers with *Plasmodium falciparum* sporozoites, attenuated by radiation such that they can invade the host hepatocyte but not develop to the blood stage form, can confer sterile protective immunity against challenge with infectious, non-attenuated sporozoites. Protection is mediated by T cell responses directed against antigens expressed in the infected hepatocyte.

Our strategy is to incorporate multiple, pre-erythrocytic antigens into a single, adenovector vaccine to target the liver stage of the *P. falciparum* life cycle. We have chosen to use mammalian codon-optimized forms of *Pf*CSP, *Pf*LSA1, and *Pf*Ag2 as the pre-erythrocytic antigens for inclusion in our vaccine. In animal models as well as in humans, all three antigens are known to induce T cell responses that are thought to be associated with protective immunity, and vaccines based on *Pf*CSP and *Pf*Ag2 have conferred sterile protection against sporozoite challenge in animals.

Construction of multivalent adenovectors requires multiple promoters, as redundant use of the hCMV promoter would lead to rearrangements in the adenovector genome. We therefore conducted a systematic evaluation of promoters suitable for driving antigen expression in an E1/E3/E4 deleted adenovector for capacity to induce antigen-specific T cell and antibody responses and protective immunity in mice. We demonstrated a threshold level of promoter activity capable of inducing robust antigen-specific T cell and antibody responses. We identified multiple promoters that expressed antigens above this threshold and generated adenovectors utilizing these promoters to express one, two, or all three antigens. We explored multiple sites and configurations of these expression cassettes in the adenovector genome and identified configurations that express all antigens efficiently. We further demonstrated that the bivalent adenovectors induced robust T cell and antibody responses in mice against both antigens, with no apparent antigen interference. We are currently evaluating the trivalent pre-erythrocytic stage adenovectors in mouse immunogenicity experiments and these data are forthcoming. Our data indicate that multiple antigen expression adenovectors may be a cost-effective way to induce and broaden protective immunity against malaria.

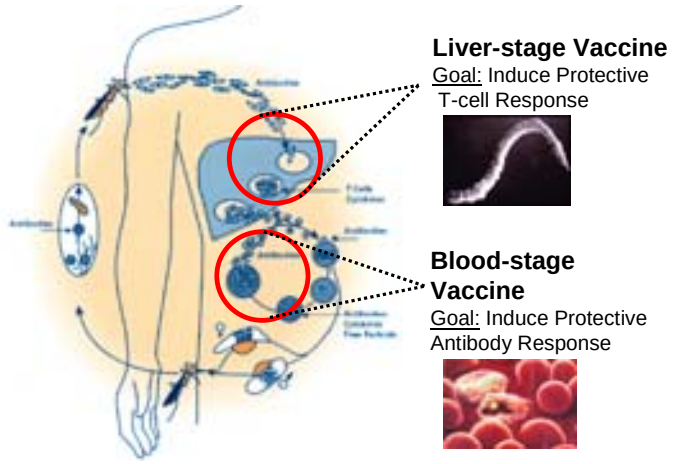
Liver-stage Vaccine
Goal: Induce Protective T-cell Response
Blood-stage Vaccine
Goal: Induce Protective Antibody Response

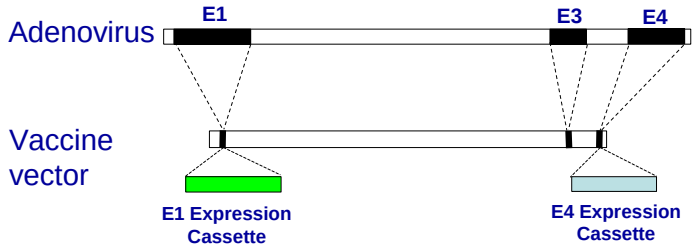
Adenovirus
E1
E3
E4
Vaccine vector
E1 Expression Cassette
E4 Expression Cassette

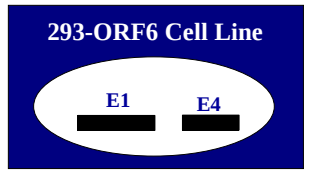
293-ORF6 Cell Line
E1
E4

Advantages of the 293-ORF6 cell line

- Robust RCA free production of E1/E3/E4-deleted adenovectors
- Extensively characterized cell line
- Biological Master File at FDA
- Serum free bioreactor process
- Efficient growth of alternative serotype vectors
- Potential for production of other viral vectors

Rationale for Multivalent Adenovector

- Increased breadth of antigen specific immune response by:
 - Expressing multiple antigens
 - Expressing antigens and immune stimulators
- Reducing the cost of the vaccine (critical in the developing world)
- Increasing potency

Requirements of Multivalent Adenovector

- Sufficient capacity to insert multiple antigens and expression cassettes
- Sufficient antigen expression to induce appropriate immune responses
 - Multiple promoters and poly A sequences to prevent vector rearrangements
 - Multiple locations in genome for insertion of antigen expression cassettes
- High Vector Yields
- Genetic stability

A

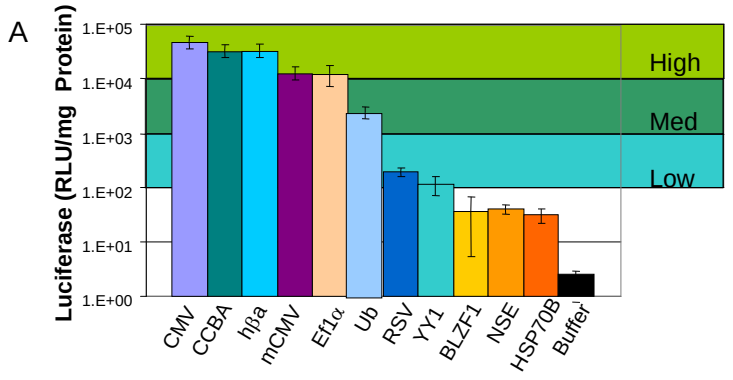
Luciferase (RLU/mg Protein)
1.E+05
1.E+04
1.E+03
1.E+02
1.E+01
1.E+00
High
Med
Low
CMV
CCBA
hβa
mCMV
Ef1α
Ub
RSV
YY1
BLZF1
NSE
HSP70B
Buffer

B

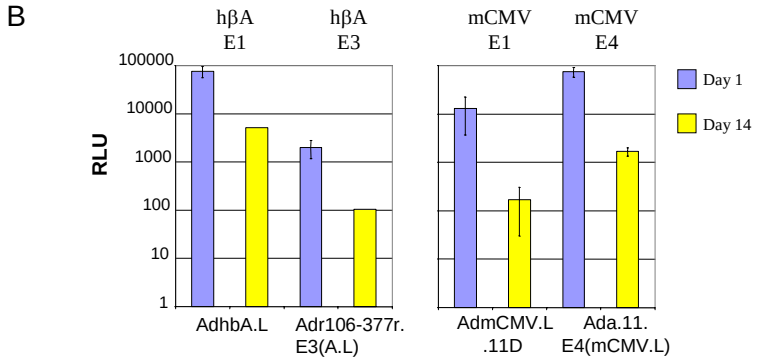
hβA E1
hβA E3
mCMV E1
mCMV E4
RLU
100000
10000
1000
100
10
1
Day 1
Day 14
AdhbA.L
Adr106-377r. E3(A.L)
AdmCMV.L .11D
Ada.11. E4(mCMV.L)

Figure 1. Relative Promoter Strength *in vivo*. Cohorts of five female C7Bl/6 mice were injected with 1e10 viral particles into the tibialis muscle as used in the vaccination studies, harvested 24 hours later and luciferase levels measured in relative light units (RLU)/ug protein. (A) Vectors: E1 region replaced with an expression cassette and contain deletions of their E3 and E4 regions. The promoters assayed were: CMV=human CMV, CCBA= CMV enhancer + Chicken Beta Actin chimeric promoter, hβA=human beta actin, mCMV=murine CMV, Ef1α=Elongation factor alpha, Ub=Ubiquitin, RSV=Rous Sarcoma Virus, YY1=Ying Yang 1, BLZF1=BLZF1, NSE=Neuron Specific Enolase, HSP70B=Heat Shock Protein 70B. Buffer is a negative control. (B) Comparison of transgene expression from identical promoters positioned at different positions in the adenoviral genome.

Conclusions:

- **Several promoters have been identified that induce high levels of transgene expression *in vivo***
- **High level expression can be achieved from expression cassettes in the E1 and E4 regions but not from the E3 region of the adenovirus genome**

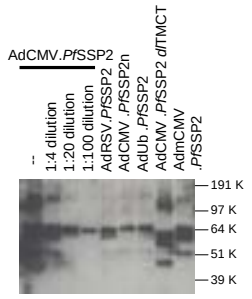
AdCMV.PfSSP2
--
1:4 dilution
1:20 dilution
1:100 dilution
AdRSV.PfSSP2
AdCMV .PfSSP2n
AdUb .PfSSP2
AdCMV .PfSSP2 dlTMCT
AdmCMV .PfSSP2
191 K
97 K
64 K
51 K
39 K

Figure 2. Adenovector mediated expression of *Pf*SSP2 from various promoters *in vitro*. Human embryonic lung (HEL) cells at 80% confluence were infected with 100 pu/cell with the indicated vectors expressing *Pf*SSP2. 24 hours later the media was removed, the plates were washed 3 times with PBS, and the cells were lysed in 1 ml RIPA buffer. 20ul of the lysates were electrophoresed on a 10% polyacrylamide gel, transferred to a nylon membrane and the *Pf*SSP2 protein products were identified using a *Pf*SSP2.1 specific antibody.

A

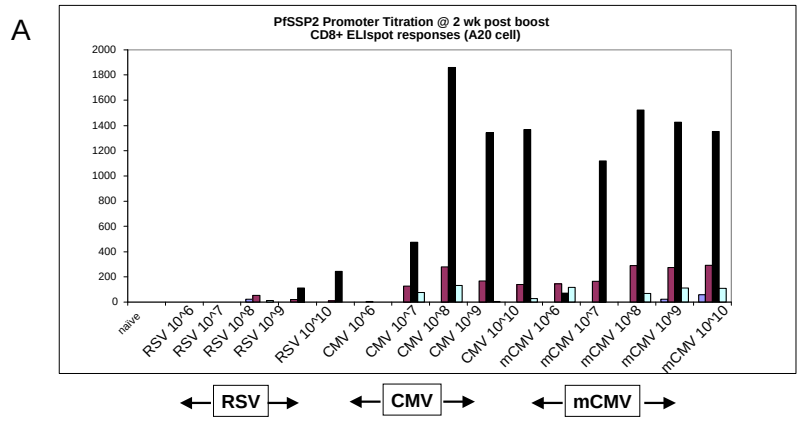
PfSSP2 Promoter Titration @ 2 wk post boost
CD8+ ELIspot responses (A20 cell)
2000
1800
1600
1400
1200
1000
800
600
400
200
0
naïve
RSV 10^6
RSV 10^7
RSV 10^8
RSV 10^9
RSV 10^10
CMV 10^6
CMV 10^7
CMV 10^8
CMV 10^9
CMV 10^10
mCMV 10^6
mCMV 10^7
mCMV 10^8
mCMV 10^9
mCMV 10^10
RSV
CMV
mCMV

B

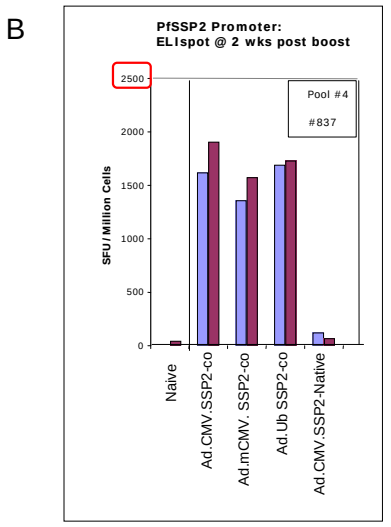
PfSSP2 Promoter:
ELIspot @ 2 wks post boost
SFU / Million Cells
2500
2000
1500
1000
500
0
Pool #4
#837
Naive
Ad.CMV.SSP2-co
Ad.mCMV. SSP2-co
Ad.Ub SSP2-co
Ad.CMV.SSP2-Native

Figure 3. Immunogenicity of adenovectors expressing various forms of *Pf*.SSP2 from different promoters. BALB/c mice received intramuscular immunizations of adenovector. T cell responses were measured by ELIspot assays 2 weeks following the last administration of vector. (A) Dose response following a single administration of adenovector. (B) Comparison of Ubiquitin (Ub), CMV and mCMV promoters and native to codon optimized antigen following two administrations of 1e8 pu of adenovector at a 6 week interval.

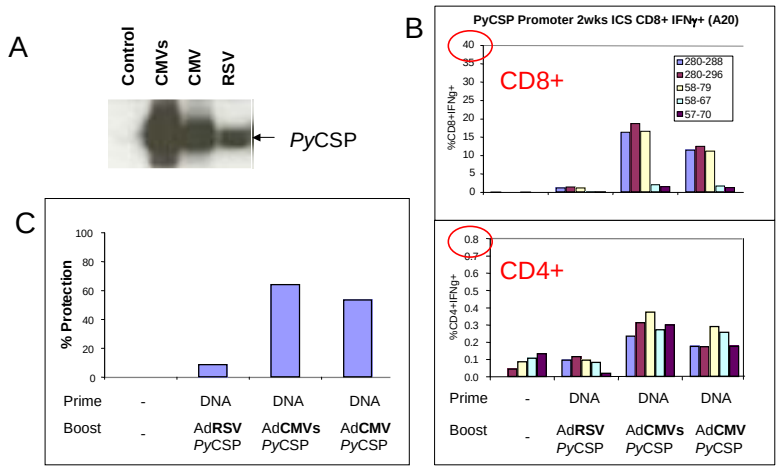
A
Control
CMVs
CMV
RSV
PyCSP
B
PyCSP Promoter 2wks ICS CD8+ IFNγ+ (A20)
CD8+
%CD8+IFNg+
280-288
280-296
58-79
58-67
57-70
CD4+
%CD4+IFNg+
Prime
Boost
DNA
AdRSV PyCSP
AdCMVs PyCSP
AdCMV PyCSP
C
% Protection
100
80
60
40
20
0

Figure 4. *In vitro* expression and immunogenicity of adenovectors expressing various forms of *Py*CSP from different promoters. (A) *Py*CSP expression by immunoblot assay following infection of HEL cells. BALB/c mice were primed by intramuscular immunizations of DNA followed by adenovector boost. (B) T cell responses were measured by ICS 2 weeks following the last administration of adenovector. (A) Dose response following a single administration of adenovector. (B) Protection from challenge by *P.yoelii* sporozoites assayed by analysis of blood smears taken at various times post challenge.

Table 1. Summary of expression and immunogenicity data of adenovectors expressing various forms of *Pf*.SSP2 from different promoters.

Promoter	Antigen	Predicted Expression	Expression in vitro	T Cell Responses	Antibody Responses
Human CMVs	SSP2	++++	++++	++++	++++
Human CMVs	SSP2 dlTS	++++			
Human CMVs	SSP2 dlTMCT	++++	++++	++++	++++
Murine CMV	SSP2	+++	+++	++++	++++
Ubiquitin	SSP2	++	+	++++	++++
Human CMVs	SSP2 (native)	+	+	+	+
RSV	SSP2	+	+	+	+

Bivalent Pre-erythrocytic Stage Adenovector

A. Schematic view of bivalent adenovector genome

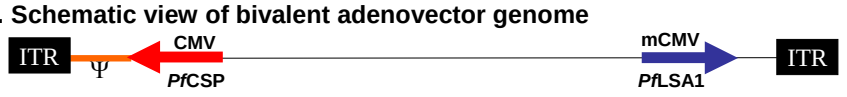
ITR
Ψ
CMV
PfCSP
mCMV
PfLSA1
ITR

B. Bivalent adenovector both *Pf*CSP and *Pf*LSA1 antigens

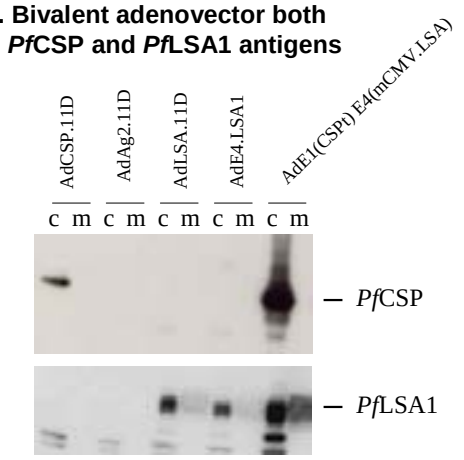
AdCSP.11D
AdAg2.11D
AdLSA.11D
AdE4.LSA1
AdE1(CSPt) E4(mCMV.LSA)
c m c m c m c m c m
PfCSP
PfLSA1

C. Adenovector induces potent T cell responses to both antigens

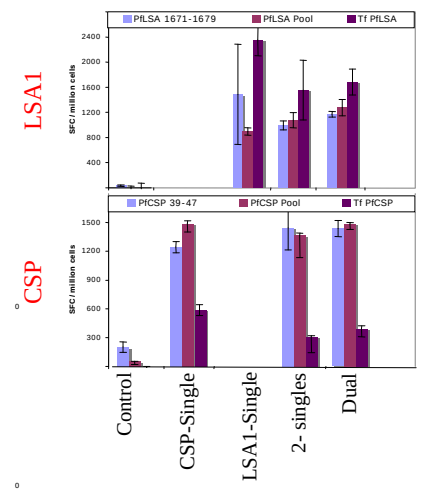
LSA1
PfLSA 1671-1679
PfLSA Pool
Tf PfLSA
CSP
PfCSP 39-47
PfCSP Pool
Tf PfCSP
SFC/million cells
Control
CSP-Single
LSA1-Single
2- singles
Dual

D. Bivalent adenovector induces potent antibody responses in mice

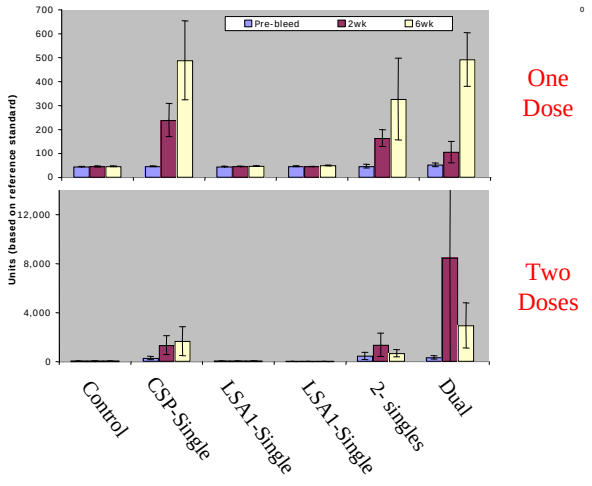
Pre-bleed
2wk
6wk
Units (based on reference standard)
700
600
500
400
300
200
100
0
12,000
8,000
4,000
One Dose
Two Doses
Control
CSP-Single
LSA1-Single
LSA1-Single
2- singles
Dual

Figure 5. Antigen expression and immunogenicity of bivalent adenovector expressing *Pf*CSP and *Pf*LSA. (A) Schematic representation of bivalent adenovectors (B) *Pf*CSP and *Pf*LSA1 expression by immunoblot assay following infection of HEL cells, c and m represent cell lysate and media. (C) BALB/c mice were immunized by intramuscular immunizations of adenovector and T cell responses were measured by ELIspot 2 weeks following vector administration. (D) Antibody responses to *Pf*CSP were measured by ELISA at various times post administration.

Conclusions: Multivalent Pre-erythrocytic Stage Adenovector Vaccine

- **Promoters**
 - **Threshold levels of antigen expression required for strong antibody and T cell responses**
 - **Identified several promoters that drive expression at or beyond this threshold level**
 - **E1 and E4 regions are preferred for antigen expression**
- **Bivalent vector**
 - **Strong T cell and antibody responses to both antigens**